Abstract no. 8104

Safety, tolerability, and preliminary efficacy results from an ongoing Phase 1 study of LB2102, a dnTGFBR2-armored DLL3-targeted autologous CAR-T cell therapy, in patients with relapsed or refractory SCLC or LCNEC

Jacob Sands MD[1]; Alberto Chiappori MD[2]; Ben Creelan MD[2]; Paul Schwarzenberger MD[3]; Mythili Koneru MD PhD[3]; Sahista Vahora PharmD[3]; Christian Davis MS[3]; Da Xu PhD[3]; Chuan Wang PhD[3]; Reinhold Munker MD[4]; Zhonglin Hao MD PhD[4]; and Adam J. Schoenfeld MD[5]

email : jacob_sands@dfci.harvard.edu

[1]Dana Farber Cancer Institute, Boston, MA; [2]Moffitt Cancer Center, Tampa, FL; [3]Legend Biotech USA Inc, Somerset, NJ; [4]Markey Cancer Center, Lexington, KY; [5]Memorial Sloan Kettering Cancer Center, New York City, NY

ClinicalTrials.gov no: NCT05680922

Introduction

Delta-like ligand 3 (DLL3) is a promising therapeutic target for small cell lung carcinoma (SCLC) and other neuroendocrine tumors. We present preliminary results from the phase 1, dose-escalation study of LB2102, an autologous CAR-T cell therapy engineered to target DLL3 armored with a TGF-beta receptor(TGFBR) blockade to overcome the immunosuppressive tumor microenvironment.

Methods

- Open-label, multicenter, phase 1 study evaluating one-time treatment with LB2102 in patients with SCLC or large cell neuroendocrine carcinoma (LCNEC) who have relapsed after or are refractory to ≥1 prior line of therapy.
- LB2102 is administered via single intravenous infusion after lymphodepleting chemotherapy.
- Dose escalation follows a modified 3+3 design, with planned dose levels (DLs) of 0.3, 1.0, 2.0, 4.0, 8.0, 12.0, 16.0 $\times 10^6$ CAR+ T cells/kg. Data from only DLs 1-4 are presented.
- All subjects underwent 3-day lymphodepletion with fludarabine (30 mg/m^2), and cyclophosphamide (300 mg/m^2).

Primary Objective: to assess the safety, tolerability and determine the recommended phase 2 dose (RP2D).

Secondary objectives: to evaluate the efficacy, pharmacokinetics (PK) and immunogenicity of LB2102.

Study Population:

- ≥ 18 years old with histologically/cytologically confirmed LCNEC and/or SCLC
- Progression or insufficient response after ≥1 prior line of treatment
- ECOG of 0 or 1, life expectancy of >4 months
- ≥1 lesion measurable by radiology, per RECIST v.1.1
- Adequate organ function per protocol

Key Exclusion Criteria

- Prior cellular or DLL3-targeted therapy
- History of checkpoint inhibitor-associated pneumonitis
- Ascites, pleural/peritoneal effusions
- Leptomeningeal or active/symptomatic brain metastases. Treated brain metastases are allowed given disease stabilization after definitive therapy
- Active autoimmune disease treatment with immunomodulators
- Other disorder that could pose safety risk or interfere with study results or compliance

Acknowledgements: This study was funded by Legend Biotech USA Inc. and Novartis AG. We would like to thank the patients, their families, and caregivers for their contribution to this research.

Baseline Demographics and Characteristics

- As of 25 April 2025, 12 patients received infusions LB2102 at DL1 0.3 $\times 10^6$ (n=3), DL2 1×10^6 (n=3), DL3 2×10^6 (n=3), and DL4: 4.0 × 10^6 CAR+ T cells/kg (n=3). Dose escalation is ongoing and could go to DL7.
- All patients had either SCLC or LCNEC with metastases, most patient received bridging therapy (n=11).

Patient Demographics, Baseline Characteristics, and Disease Characteristics at Screening	DL1: 0.3 × 10^6 CAR+ T cells/kg [N=3]	DL2: 1.0 × 10^6 CAR+ T cells/kg [N=3]	DL3: 2.0 × 10^6 CAR+ T cells/kg [N=3]	DL4: 4.0 × 10^6 CAR+ T cells/kg [N=3]	Overall [N=12]
Age (years) at Consent					
Mean (SD)	57.3 (3.5)	41.7 (19.1)	53.0 (6.6)	61.3 (7.0)	53.3 (12.0)
Age: n (%)					
≥60 years	1 (33.3)	0	0	2 (66.7)	3 (25.0)
Sex: n (%)					
Female	1 (33.3)	1 (33.3)	2 (66.7)	3 (100)	7 (58.3)
Race and Ethnicity: n (%)					
White	3 (100)	3 (100)	2 (66.7)	3 (100)	11 (91.7)
Other	0	0	1 (33.3)	0	1 (8.3)
Hispanic or Latino	0	0	1 (33.3)	0	1 (8.3)
Primary Tumor Type: n (%)					
SCLC	3 (100)	2 (66.7)	3 (100)	2 (66.7)	10 (83.3)
LCNEC	0	1 (33.3)	0	1 (33.3)	2 (16.7)
Disease State at Initial Diagnosis: n (%)					
Extensive	3 (100)	1 (33.3)	1 (33.3)	2 (66.7)	7 (58.3)
Limited	0	1 (33.3)	0	1 (33.3)	2 (16.7)
Unknown	0	1 (33.3)	2 (66.7)	0	3 (25.0)
Histologic Grade at Initial Diagnosis: n (%)					
Poorly Differentiated	3 (100)	2 (66.7)	0	2 (66.7)	7 (58.3)
Unknown	0	1 (33.3)	3 (100)	1 (33.3)	5 (41.7)
History of Brain Metastases n (%)	2 (66.7)	3 (100)	1 (33.3)	2 (66.7)	8 (66.7)
Number of Lines of Prior Therapies					
Median (Min, Max)	1.0 (1, 2)	2.0 (2, 5)	1.0 (1, 1)	1.0 (1, 2)	1.0 (1, 5)
Prior Radiotherapies: n (%)	2 (66.7)	2 (66.7)	2 (66.7)	3 (100)	9 (75.0)
Prior Platinum-Based Therapies n(%)	3 (100)	3 (100)	2 (66.7)	3 (100)	11 (91.7)
Prior Alkylating Agents: n (%)	1 (33.3)	2 (66.7)	0	0	3 (25.0)
Other: n (%)	3 (100)	3 (100)	3 (100)	3 (100)	12 (100)
Number of Lines of Bridging Therapies					
Median (Min, Max)	1.0 (1, 1)	1.0 (1, 2)	1.0 (1, 1)	1.0 (0, 1)	1.0 (0, 2)

Safety

- There were 3 patients with treatment-emergent adverse events (TEAEs) in each dose level (12 total) (**Table 1**). One patient had a maximum grade TEAE of Grade 1 (DL4), 4 had a maximum of Grade 2 (DL1 and DL4), 3 had a maximum of Grade 3 (DL2 and DL3), and 4 had maximum of Grade 4 (DLs 2, 3, and 4). There were no deaths.
- There were 7 patients with TEAEs related to LB2102 infusion (**Table 2**). Grade ≥3 TEAEs attributed to LB2102 included anemia (n=2), leukopenia (n=2) and neutropenia (n=2); none were classified as serious, and all were deemed related to lymphodepletion.
- Cytokine-release syndrome (CRS) occurred in 2 patients (both Grade 1, at DL3 and DL4); fever was the only symptom. The CRS events occurred 6 days (DL3) and 16 days (DL4) after infusion of LB2102, with durations of 1 day and 2 days, respectively.
- There were 5 patients with serious TEAEs total (**Table 1**). One serious TEAE was related to LB2102 infusion (Grade 1 CRS).
- There were no adverse events of special interest (AESIs), dose-limiting toxicities (DLTs), neurotoxicity, or TEAEs that led to discontinuation

Table 1. TEAEs

	DL1: 0.3 x 10^6 CAR+ T cells/kg [N=3] n (%)	DL2: 1.0 × 10^6 CAR+ T cells/kg [N=3] n (%)	DL3: 2.0 × 10^6 CAR+ T cells/kg [N=3] n (%)	DL4: 4.0 × 10^6 CAR+ T cells/kg [N=3] n (%)	Overall [N=12] n (%)
Any TEAE [a]	3 (100)	3 (100)	3 (100)	3 (100)	**12 (100)**
Grade 1	0	0	0	1 (33.3)	**1 (8.3)**
Grade 2	3 (100)	0	0	1 (33.3)	**4 (33.3)**
Grade 3	0	1 (33.3)	2 (66.7)	0	**3 (25.0)**
Grade 4	0	2 (66.7)	1 (33.3)	1 (33.3)	**4 (33.3)**
Grade 5	0	0	0	0	**0**
Any Serious TEAE	1 (33.3)	1 (33.3)	1 (33.3)	2 (66.7)	**5 (41.7)**
Related to LB2102	0	0	0	1 (33.3)	**1 (8.3)**
Any AESI	0	0	0	0	**0**
Any DLT	0	0	0	0	**0**
Any TEAE Leading to follow-up discontinuation	0	0	0	0	**0**

[a] The number of patients with at least 1 TEAE is summarized at their maximum severity grade.
AESIs in this trial are CRS Grade ≥ 3, any-grade neurotoxicity, infection Grade ≥ 4, or new primary malignancy.

Table 2. TEAEs Related to LB2102 Infusion

Preferred Term	DL1: 0.3 × 10^6 CAR+ T cells/kg [N=3] n (%)		DL2: 1.0 × 10^6 CAR+ T cells/kg [N=3] n (%)		DL3: 2.0 × 10^6 CAR+ T cells/kg [N=3] n (%)		DL4: 4.0 × 10^6 CAR+ T cells/kg [N=3] n (%)		Overall [N=12] n (%)	
	Any Grade	≥Grade 3	Any Grade	≥Grade 3	Any Grade	≥Grade 3	Any Grade	≥Grade 3	Any Grade	≥Grade 3
Any TEAE related to LB2102 Infusion	2 (66.7)	0	2 (66.7)	2 (66.7)	1 (33.3)	0	2 (66.7)	0	**7 (58.3)**	**2 (16.7)**
Anemia	0	0	2 (66.7)	2 (66.7)	0	0	1 (33.3)	0	**3 (25.0)**	**2 (16.7)**
CRS	0	0	0	0	1 (33.3)	0	1 (33.3)	0	**2 (16.7)**	**0**
Hypotension	1 (33.3)	0	1 (33.3)	0	0	0	0	0	**2 (16.7)**	**0**
Nausea	2 (66.7)	0	0	0	0	0	0	0	**2 (16.7)**	**0**
Neutrophil count decreased	0	0	2 (66.7)	2 (66.7)	0	0	0	0	**2 (16.7)**	**2 (16.7)**
White blood cell count decreased	0	0	2 (66.7)	2 (66.7)	0	0	0	0	**2 (16.7)**	**2 (16.7)**
Arthralgia	0	0	0	0	1 (33.3)	0	0	0	**1 (8.3)**	**0**
Muscular weakness	0	0	0	0	0	0	1 (33.3)	0	**1 (8.3)**	**0**

Pharmacokinetics

- Significant expansion of CAR-T cells in peripheral blood was observed as measured by qPCR at DL3 (n=3) and DL4 (n=3) (**Table 3, Figure 1**)
- For DL3, the median Cmax was 694 copies/µg genomic DNA (range, 45.6-2260), with a median Tmax of 15 days (range, 10-29)
- For DL4, the median Cmax was 581 copies/µg genomic DNA (range, 564-1130 days), with a median Tmax of 15 days (range, 5-20)

Table 3. PK Parameters for CAR Transgene Copy Numbers in Blood

	DL1: 0.3 × 10^6 CAR+ T cells/kg [N=3]	DL2: 1.0 × 10^6 CAR+ T cells/kg [N=3]	DL3: 2.0 × 10^6 CAR+ T cells/kg [N=3]	DL4: 4.0 × 10^6 CAR+ T cells/kg [N=3]	Overall [N=12]
PK-evaluable patients	1	1	3	3	8
C_{max} (copies/ug gDNA) Median (Min,Max)	647 (647, 647)	2510 (2510, 2510)	694 (45.6, 2260)	581 (564, 1130)	671 (45.6, 2510)
T_{max}(day) Median (Min,Max)	28 (28, 28)	13 (13, 13)	15 (10, 29)	15 (5, 20)	15 (5, 29)
AUC_{last} (day*copies/ug gDNA) Median (Min,Max)	5620 (5620, 5620)	7010 (7010, 7010)	8870 (137, 44000)	6710 (2850, 15800)	6860 (137, 44000)

Figure 1. Blood Concentrations of CAR Transgene Copy Numbers in Individual Patients at DL3 and DL4

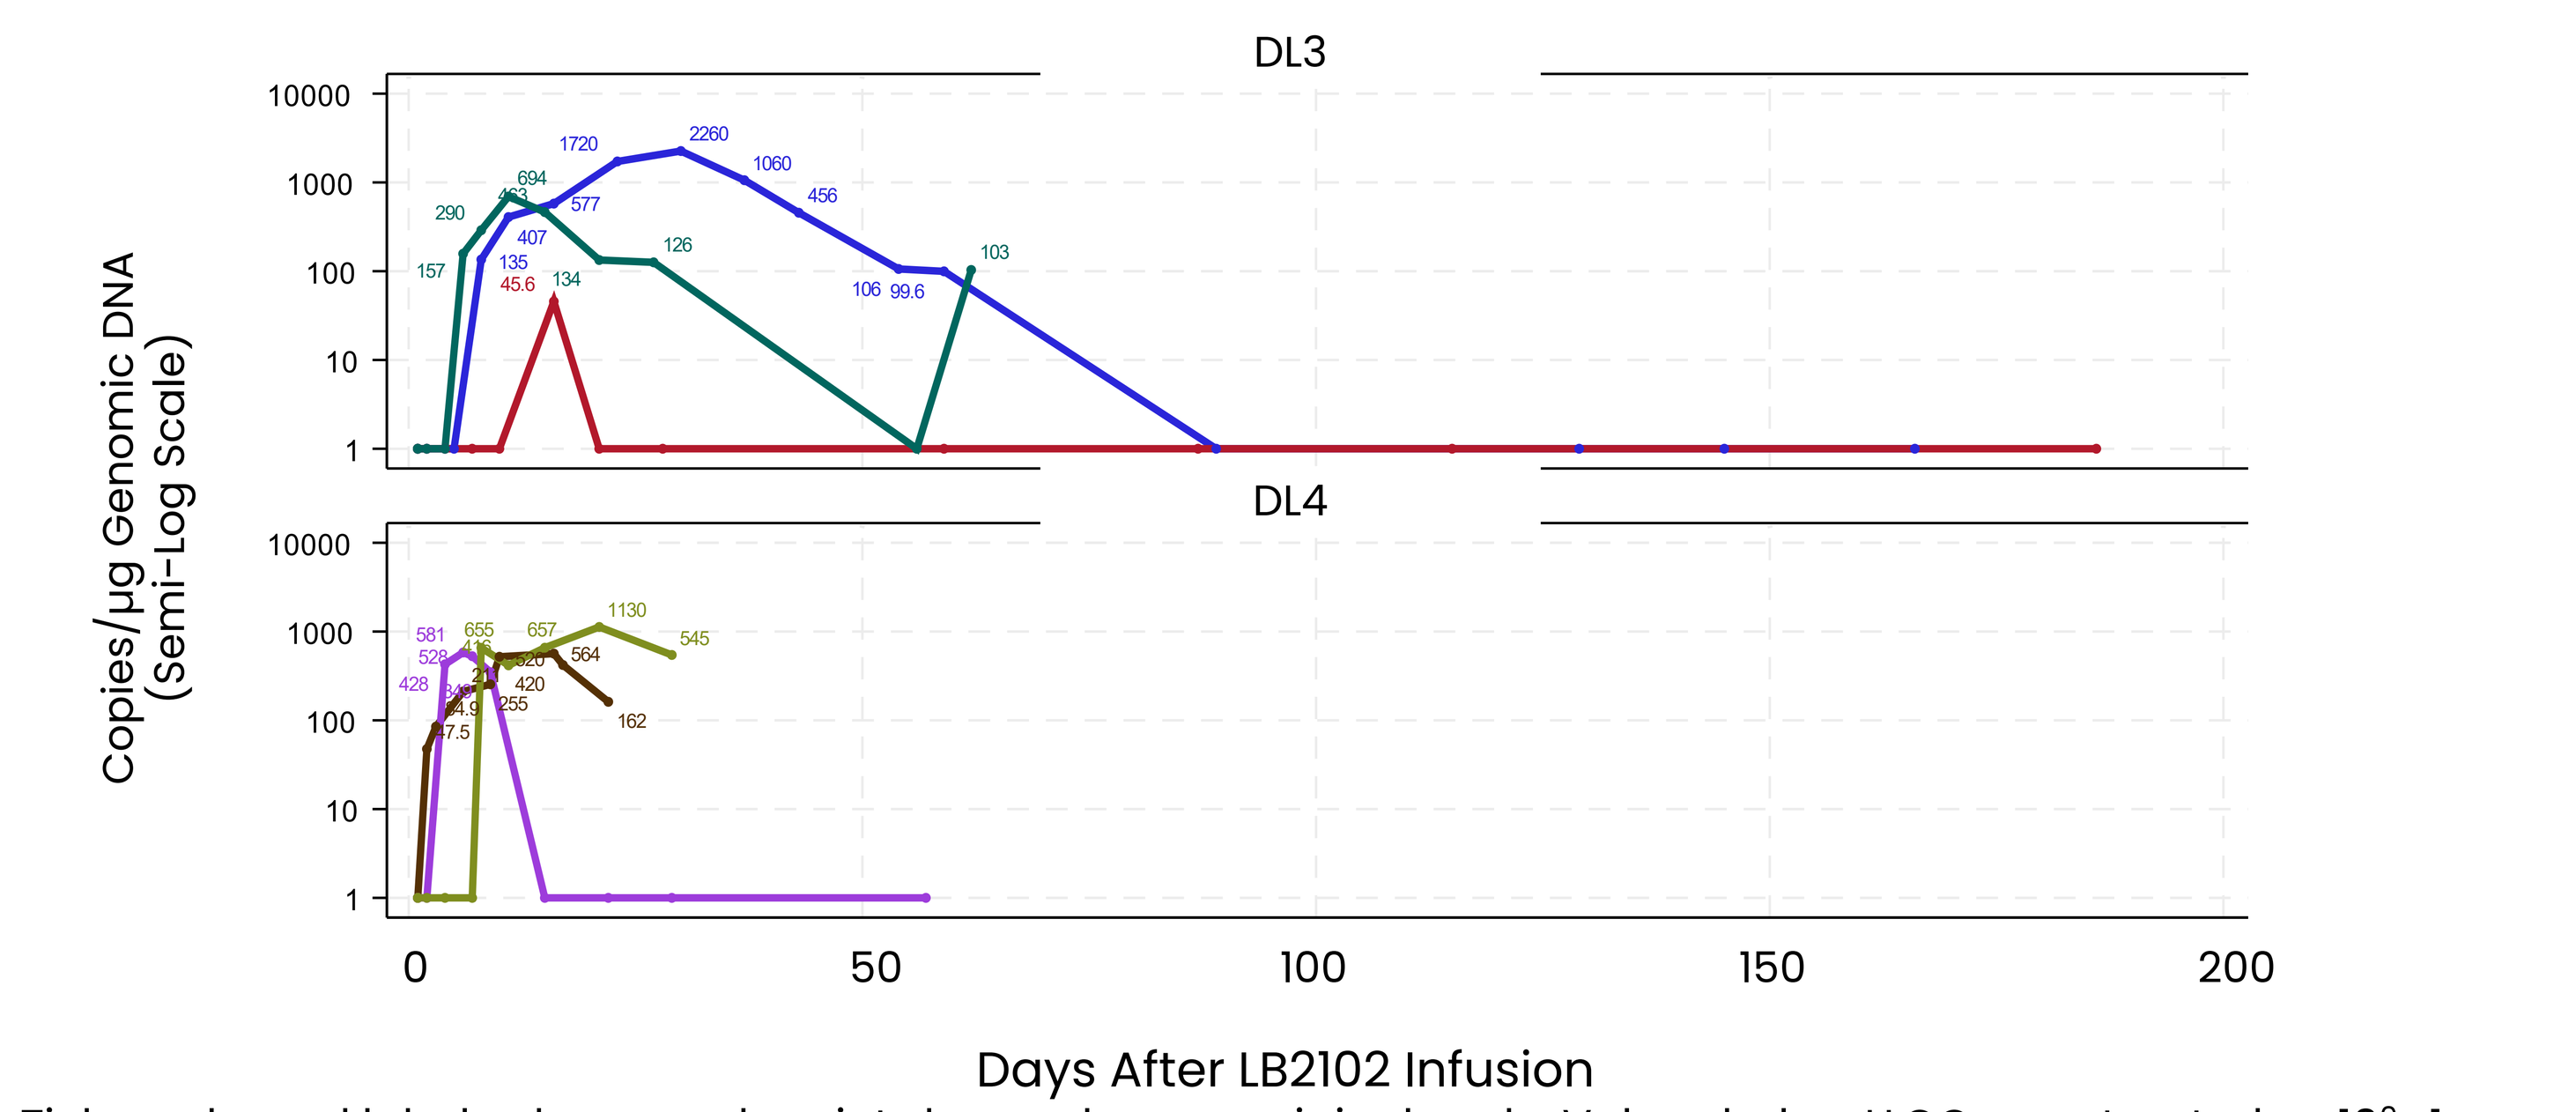

DL3
DL4
Copies/µg Genomic DNA (Semi-Log Scale)
Days After LB2102 Infusion

Tick marks and labels above each point show values on original scale. Values below LLOQ were treated as 10^0=1.

Efficacy

- The best overall responses were partial response (PR) (1 patient at DL3, 1 patient at DL4), and stable disease (SD) (3 patients at DL2, 2 patients at DL3, and 1 patient at DL4)
- The objective response rate (PR + CR) was 2/12 (16.7%)
- The disease control rate (SD + PR + CR) was 8/12 (66.7%)
- The extent of radiographic regression appeared to increase with DL; dose escalation is underway

Table 4. Patient Best Responses to LB2102

	DL1: 0.3x10^6 CAR+ T cells/kg [N=3] n (%)	DL2: 1.0x10^6 CAR+ T cells/kg [N=3] n (%)	DL3: 2.0x10^6 CAR+ T cells/kg [N=3] n (%)	DL4: 4.0x 10^6 CAR+ T cells/kg [N=3] n (%)	Overall [N=12] n (%)
Best Response (per RECIST 1.1 criteria) [a]					
CR	0	0	0	0	**0**
PR [b]	0	0	1 (33.3)	1 (33.3)	**2 (16.7)**
SD	0	3 (100)	2 (66.7)	1 (33.3)	**6 (50.0)**
Progressive disease (PD)	3 (100)	0	0	1 (33.3)	**4 (33.3)**
Disease control rate (SD+PR+CR)	0	3 (100)	3 (100)	2 (66.7)	**8 (66.7)**

[a] 1 patient had non-measurable disease at baseline on DL1
[b] 1 patient who achieved a PR with 70% tumor shrinkage also showed **no tumor metabolic activity on PET/CT**

Figure 2. Best Percent Change from Baseline in Target Lesion

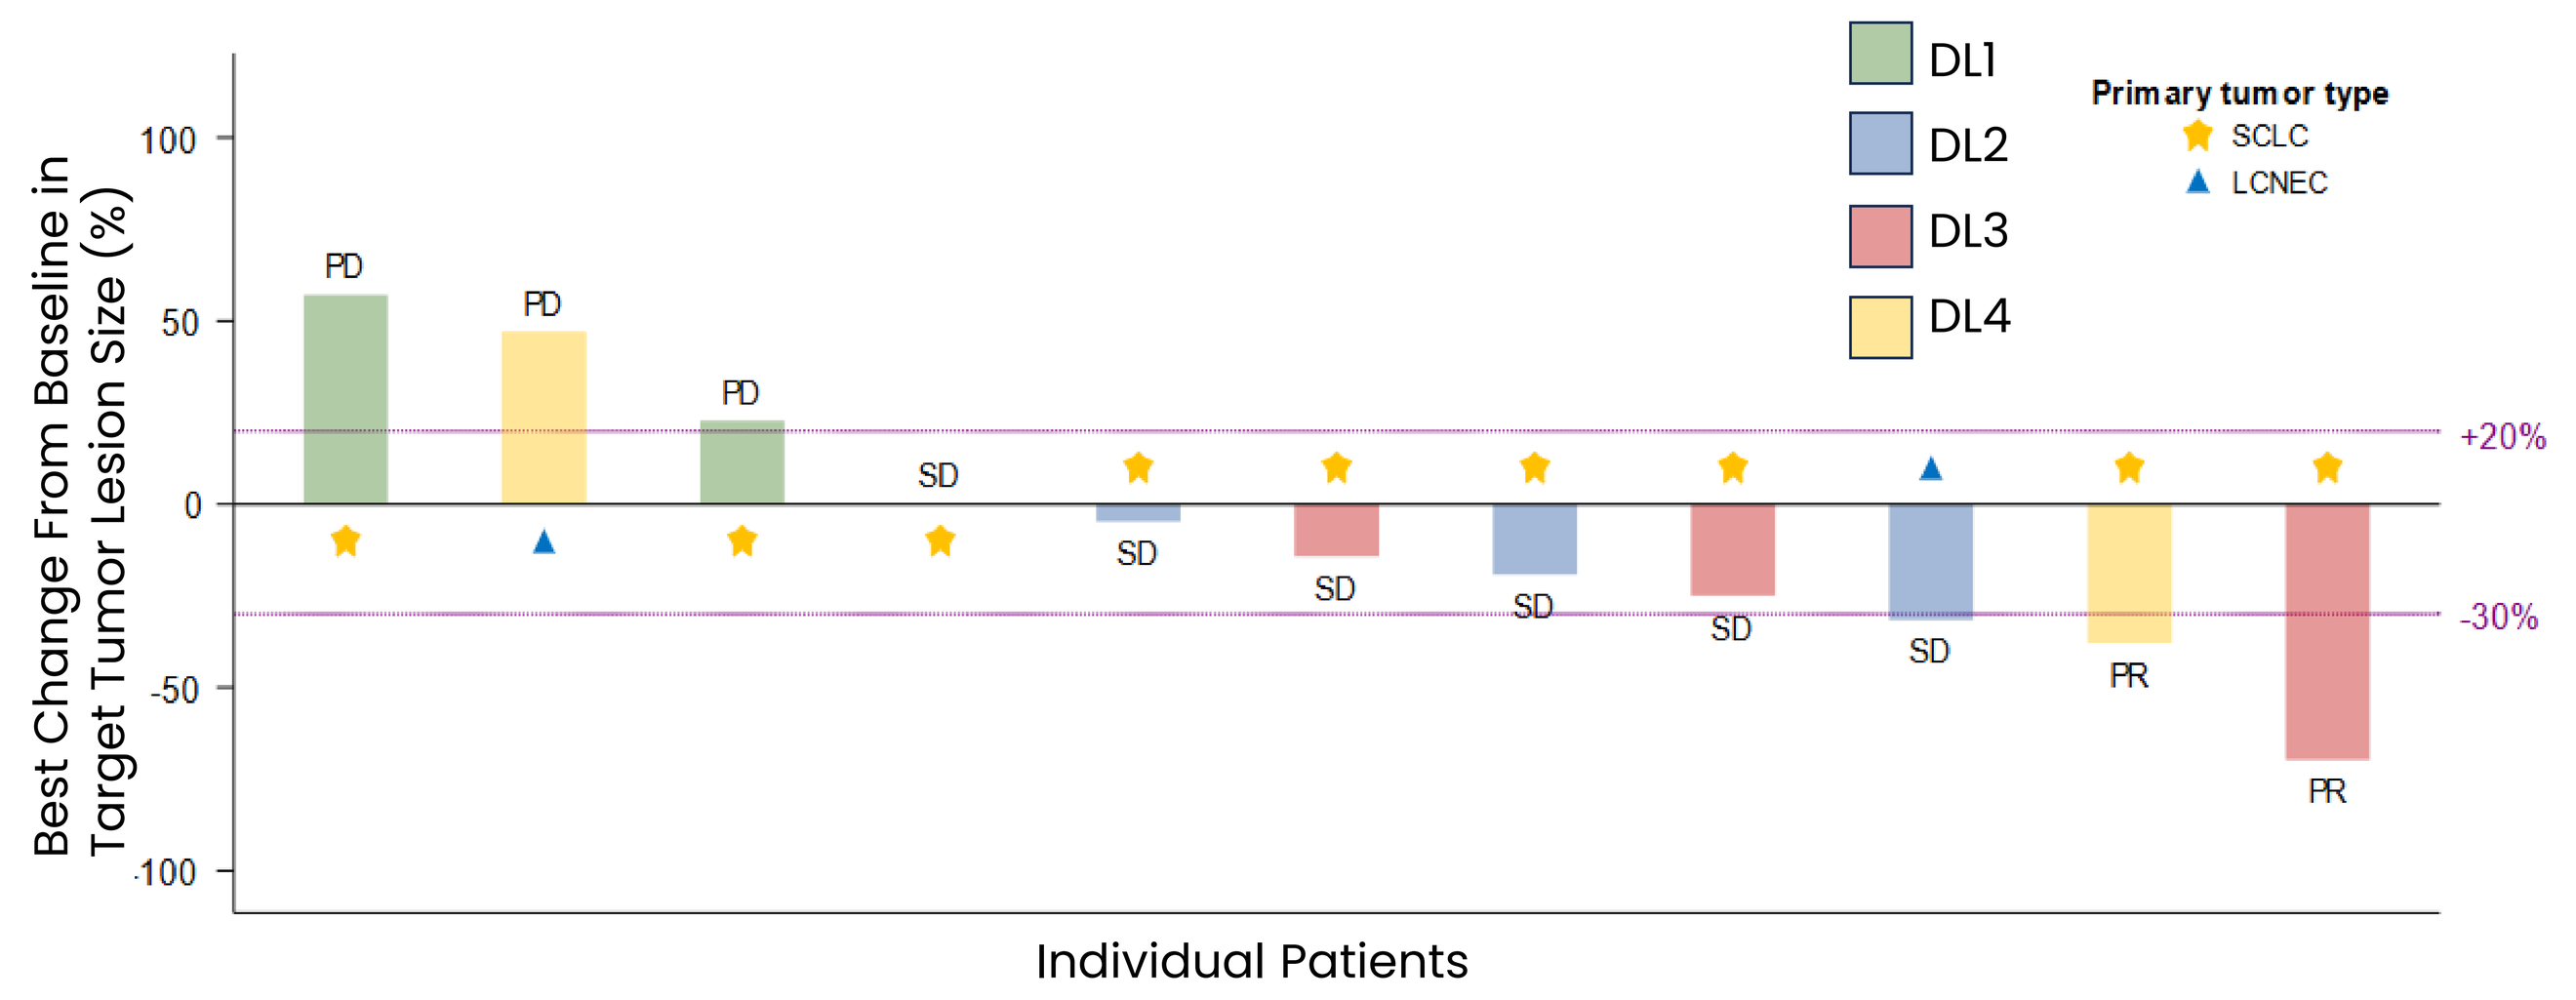

DL1
DL2
DL3
DL4
Primary tumor type
SCLC
LCNEC
Best Change From Baseline in Target Tumor Lesion Size (%)
+20%
-30%
Individual Patients

One DL1 patient progressed; not shown in the above plot due to non-measurable disease at baseline

Figure 3. Disease Response at Different Dose Levels

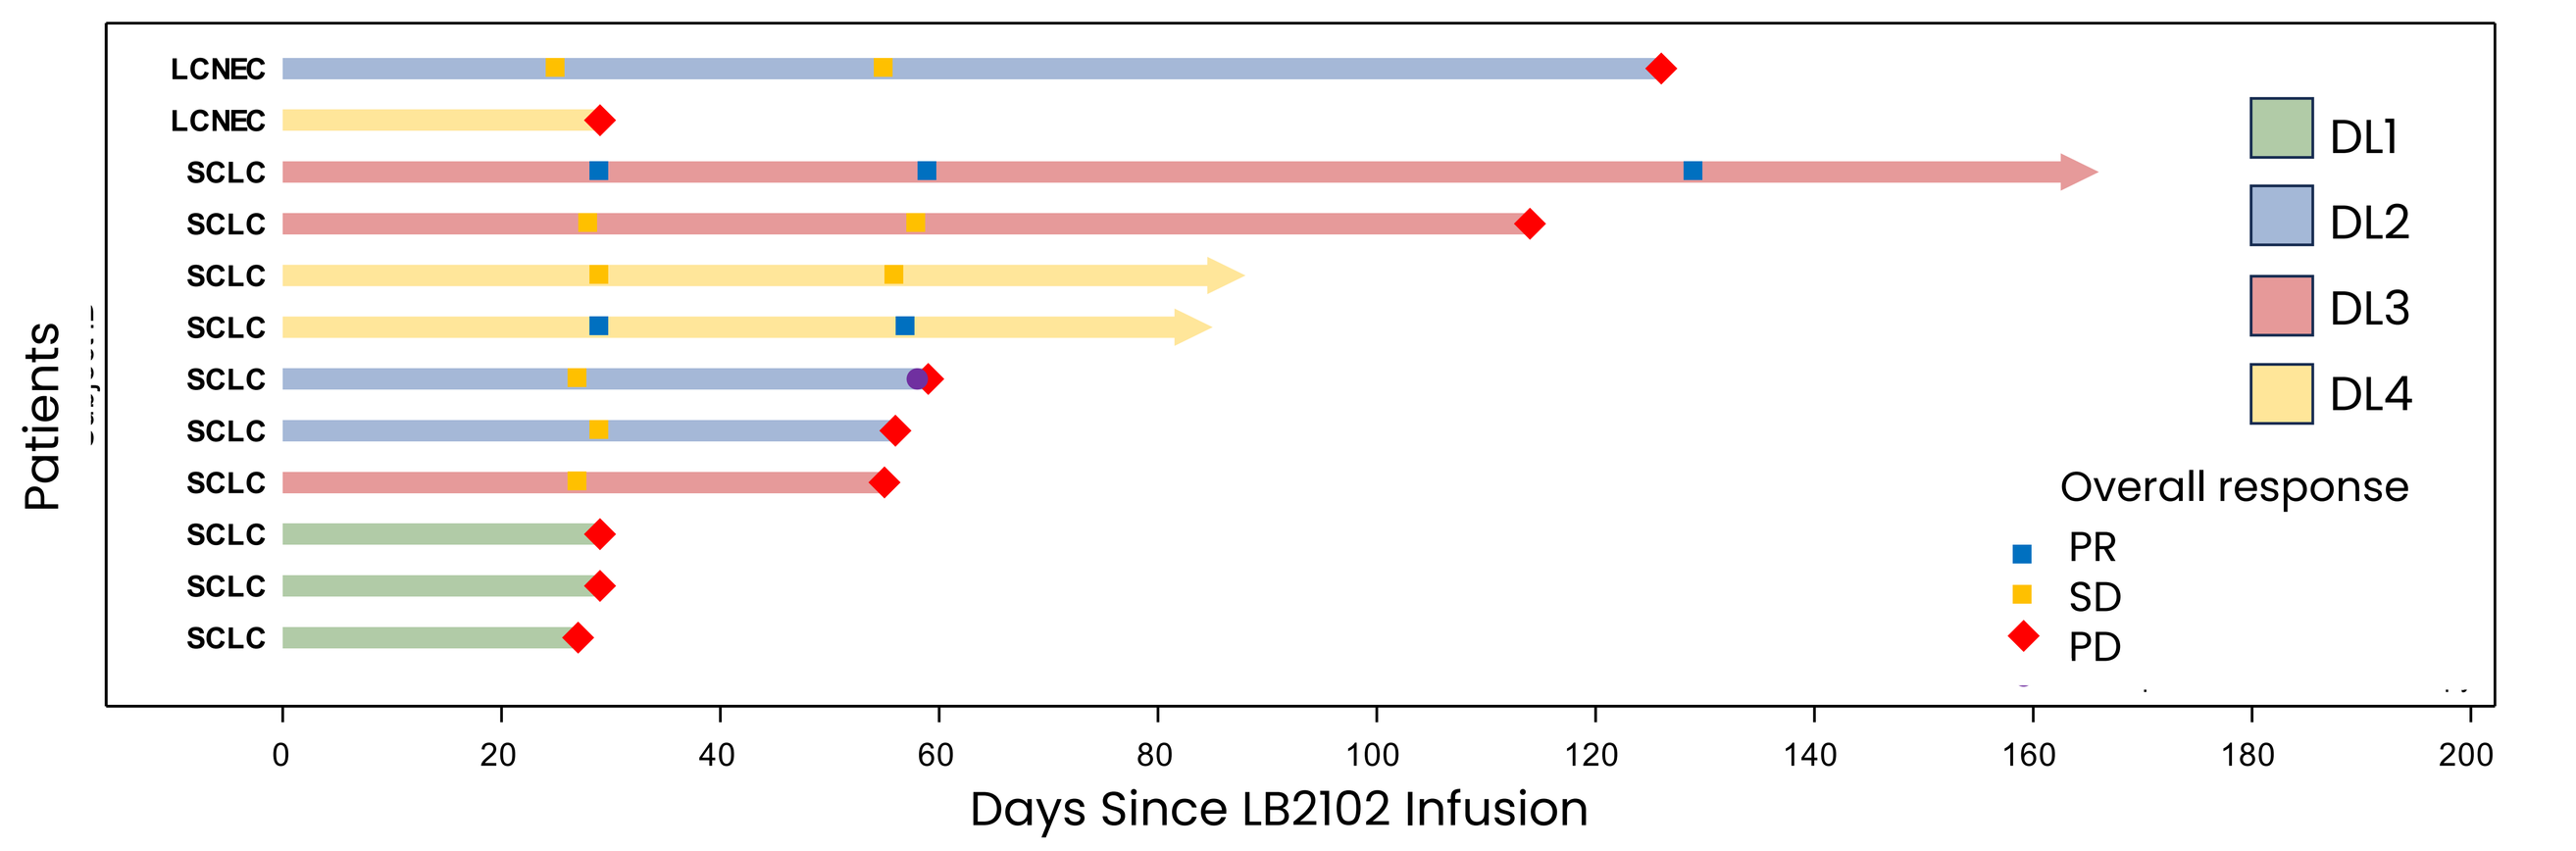

DL1
DL2
DL3
DL4
Overall response
PR
SD
PD
Patients
Days Since LB2102 Infusion

Arrowheads indicate patients (n=3) still on study follow up, without progression or anti-cancer therapy as of their last visit. The purple circle indicates when the patient received subsequent anti-cancer therapy.

Conclusions

- LB2102 has been well tolerated with no DLT observed up to DL4 (4x10^6 CAR+ T cells/kg)
- Preliminary anti-tumor activity has been observed, including a **16.7% objective response rate and 66.7% Disease Control Rate**, with responses deepening at higher dose levels
- Continued dose escalation of LB2102 in SCLC and LCNEC is warranted